950

Cabotegravir Population Pharmacokinetic Analysis of Adults & Adolescents Living with HIV or at Risk for HIV Receiving PrEP

Yu-Wei Lin,[1#] S. Y. Amy Cheung,[1#] Isabelle Deprez,[1] Susan Ford,[2] Jon Collins,[3] Cindy McCoig,[4] Conn M. Harrington,[3] Aditya Gaur,[5] Carolyn Bolton,[6] Lynda Stranix-Chibanda,[7] Sybil Hosek,[8] Mark Marzinke,[9] Brookie Best,[10] Edmund Capparelli,[10] for IMPAACT 2017 Team

[1]Certara/GSK, Princeton, NJ, USA/Collegeville, Pennsylvania, USA; [2]GSK,Collegeville, Pennsylvania, USA; [3]ViiV Healthcare, Durham, NC, USA; [4]ViiV Healthcare, Madrid, Spain; [5]St Jude Children's Research Hospital, Memphis, TN, USA; [6]Centre for Infectious Disease Research in Zambia, Lusaka, Zambia; [7]University of Zimbabwe, Harare, Zimbabwe; [8]Department of Psychiatry, Stroger Hospital of Cook County, Chicago, Illinois, USA; [9]Department of Pathology, The Johns Hopkins University, Baltimore, MD, USA; [10]University of California San Diego, San Diego, La Jolla, CA,USA

[#]Contridubed equally to this work.

Acknowledgements: *Rashmi Mehta,[2] Mark Bush,[3] Lionel Tan,[3] Kelong Han,[2] Jenny Huang,[2] Paola Coppola,[1] Kenny Watson[1], Nele Mueller-Plock[1], Sophia Fong.[1] HPTN-083-01 Team. HPTN-084-01 Team. Patients and their families.*

Introduction

- Cabotegravir (CAB) is an integrase strand transfer inhibitor approved in adults and adolescents (12 to <18 years) weighing ≥35 kg as long-acting injectable (LAI) HIV-1 prevention, and for HIV-1 treatment in combination with rilpivirine.
- An existing CAB population pharmacokinetic (PopPK) model was limited to adult PK.[1]
- We set out to extend and optimize that existing PopPK model for adolescents (12 to <18 years) by incorporating available adolescent PK data from the IMPAACT 2017/MOCHA (NCT03497676), HPTN 083-01 (NCT04692077) and HPTN 084-01(NCT04824131) clinical trials.

Methods

- The existing adult PopPK model was refined based on a new dataset, including intensive and sparse concentration, dosing, demographic, and covariate data, from adolescent participants in IMPAACT 2017, HPTN 083-01, and HPTN 084-01 (Table 1).
- All drug concentrations were measured via LC-MS/MS by a single laboratory (Clinical Pharmacology Analytical Laboratory).

Table 1. Dataset for PopPK Model Refinement

Study	Total number of participants included in the PK analysis	Number of observations available for the PK analysis
Adult Studies	1647	23389
IMPAACT 2017	147	1634
HPTN 083-01	9	88
HPTN 084-01	53	568

- The impact of the addition of CAB concentration data from IMPAACT 2017, HPTN 083-01, and HPTN 084-01 on the estimates of the PK parameters and covariate effects was assessed.
- A nonlinear mixed effects model was developed using NONMEM 7.3 using the Laplacian method.
- Individual exposure metrics were derived using empirical Bayes estimates and compared between adolescents and adults.
- Monte Carlo simulation was performed using the updated PopPK model to simulate plasma concentration versus time profiles of CAB in adult and adolescent participants over an observation period of approximately 1 year.

Results

- PK data following oral lead-in (30 mg QD, for at least 4 weeks) and LAI treatment (consisting of initial loading dose of 600 mg followed 4 weeks later by start of 400 mg Q4W or 600 mg Q8W) from 147 adolescents living with HIV (IMPAACT 2017) and 62 HIV uninfected adolescents (HPTN 083/084-01) with weight of 35.2-167 kg, BMI of 15.8-51.6 kg/m^2, and 12 to 17 years of age were added to adult data (N=1647) (Table 2).

Table 2. Baseline Characteristics

	Adult studies (N=1647)	HPTN 083-01 (N=9)	HPTN 084-01 (N=53)	IMPAACT 2017 (N=147)	Overall adolescents (N=209)	Overall population (N=1856)
Female	424 (25.7%)	0 (0%)	53 (100%)	75 (51.0%)	128 (61.2%)	552 (29.7%)
Race: Non-White	545 (33.1%)	4 (44.4%)	53 (100%)	145 (98.6%)	202 (96.7%)	747 (40.2%)
HIV uninfected participants	458 (27.8%)	9 (100%)	53 (100%)	0 (0%)	62 (29.7%)	520 (28.0%)
Participants with HIV at baseline	1189 (72.2%)	0 (0%)	0 (0%)	147 (100%)	147 (70.3%)	1336 (72.0%)
Adolescents	0 (0%)	9 (100%)	53 (100%)	147 (100%)	209 (100%)	209 (11.3%)
BMI (kg/m^2), Median [min, max]	25.4 [15.3, 69.5]	21.6 [20.0, 51.6]	22.2 [15.8, 34.1]	19.4 [16.0, 33.9]	20.4 [15.8, 51.6]	24.7 [15.3, 69.5]
Weight (kg), Median [min, max]	76.6 [41.2, 168]	70.6 [63.0, 167]	55.5 [39.9, 80.8]	47.7 [35.2, 98.5]	50.8 [35.2, 167]	74.8 [35.2, 168]

Notes: For continuous covariates, numeric columns are formatted as median [min, max]. For categorical covariates, numeric columns are formatted as count (%). N = number of participants with at least one PK observation.

Given the similarity of CAB PK between adolescents and adults, no dose adjustment as compared to the current adult label for CAB is recommended for adolescent participants.

CAB PopPK Model

- The PK of CAB following oral and LAI administration in adults and adolescents was adequately described by a 2-compartment model with first-order absorption and first-order elimination.
- All parameters were estimated with good precision, with the % RSE consistently ≤30% (Table 3).
- No new covariates were identified or added as compared to the adult PopPK model.
- Weight and smoking status were significant determinants of CL/F, and only weight was a determinant of Vc/F, Vp/F, and Q/F. Needle length, splitting of the injection, female sex, and time-varying BMI were significant determinants of Ka2 in the pooled model.
- All adolescents were non-smokers and the majority of the adolescent participants received non-split injections. Their impact on adolescent PK were assumed to be the same as the adult population.
- For the weight range 35.2-168.3 kg, CL/F and Q/F ranged from approximately 40.0% lower to 73.3% higher than for a 74.8 kg participant, and Vc/F and Vp/F ranged from approximately 43.3% lower to 84.3% higher than for a 74.8 kg participant.
- Females had 50.8% lower Ka2 values than males.
- The findings were consistent with the conclusion of the previous analyses in healthy adult participants and adult participants living with HIV.[2,3]

Table 3. Parameter Estimates and Standard Errors From the Final CAB PopPK Model

Parameter	Estimate (%RSE)	IIV (%CV)	Shrinkage (%)
Apparent clearance, CL/F (L/h)	0.145 (0.829)	23.8	10.8
Apparent central volume of distribution Vc/F (L)	5.15 (2.01)	19.0	30.6
Absorption rate constant for oral tablet, Ka1 (1/h)	1.38 (4.54)	108	69.4
Apparent intercompartmental clearance, Q/F (L/h)	0.481 (6.91)		
Apparent peripheral volume of distribution, Vp/F (L)	2.32 (5.09)		
Absorption rate constant for long-acting IM formulation, Ka2 (1/h)	0.000730 (2.24)	62.2	16.6
Relative bioavailability, F1 (-)	0.745 (0.912)	19.0	36.8
Weight on CL/F and Q/F (-)	0.676 (4.03)		
Weight on Vc/F and Vp/F (-)	0.752 (5.73)		
Current smoking status on CL/F (-)	0.193 (8.44)		
BMI on Ka2 (-)	-0.823 (10.8)		
Needle length on Ka2 (-)	0.539 (30.4)		
Gender (if female) on Ka2 (-)	-0.509 (3.72)		
Split on Ka2 (-)	0.495 (13.5)		
Additive error (µg/mL)	0.0313 (19.4)		6.80
Proportional error (-)	0.277 (1.06)		

The reference population for CL/F is a 74.8 kg non-smoker participant. The reference population for Vc/F, Vp/F, and Q/F is a 74.8 kg participant. The reference population for Ka2 is a male participant (BMI=24.8 kg/m^2) using a needle length of 1.5 inches with unsplit injection.
Abbreviations: BMI=body mass index; CI=confidence interval; IIV=inter-individual variability; IM=intramuscular; RSE=relative standard error.

Model Qualification

- The final PopPK model was able to predict the observed median and the 5th and 95th percentiles of observed CAB concentrations with reasonable accuracy (Figure 1).
- The median concentration was fully captured throughout the profile for CAB concentrations from IMPAACT 2017 (MOCHA), HPTN 083-01, and HPTN 084-01.

Figure 1. Prediction-corrected Visual Predictive Check (pcVPC) of the Final CAB PopPK Model Stratified by Study Group

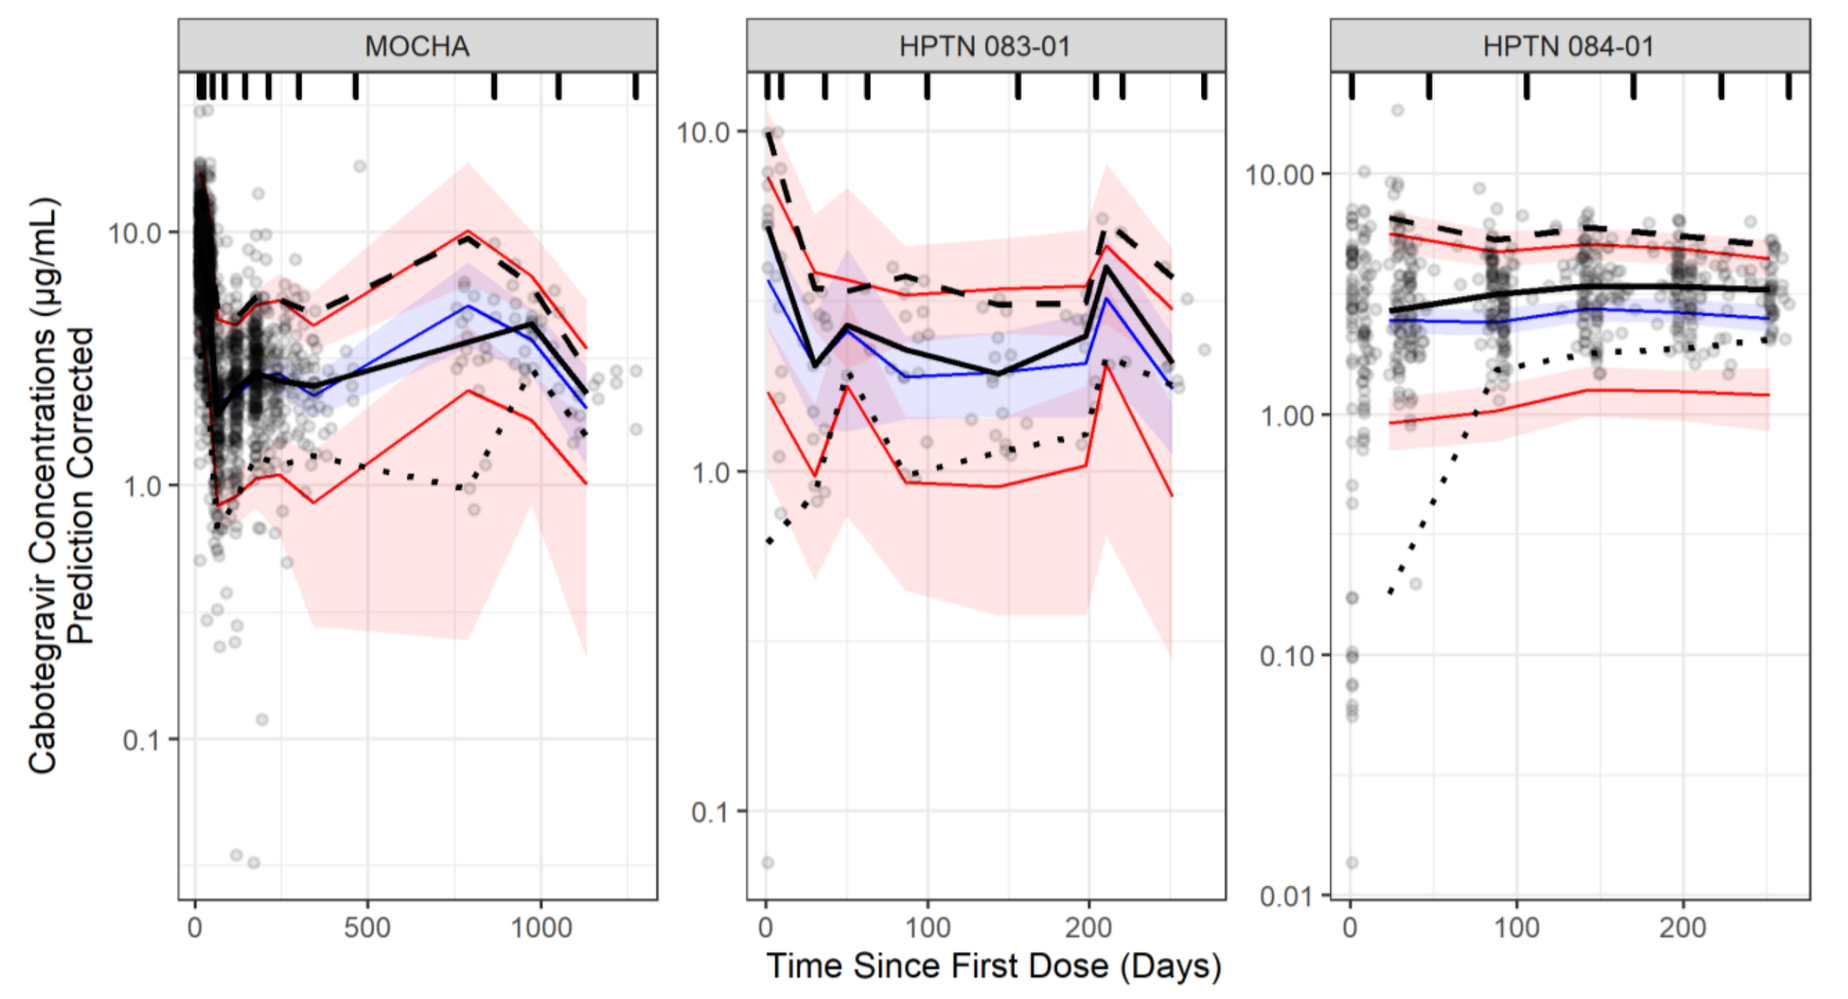
MOCHA
HPTN 083-01
HPTN 084-01
Cabotegravir Concentrations (µg/mL) Prediction Corrected
Time Since First Dose (Days)
Observed Percentiles (black lines)
5%
50%
95%
Simulated Percentiles Median (lines) 95% CI (areas)

Notes: The open circles represent the observed data. The solid black lines represent the median of the observed data, and the dashed lines represent the 5th and 95th percentiles of the observed data. The blue solid lines represent the median of the simulated data, and the pink solid lines represent the 5th and 95th percentiles of the simulated data. The blue and pink shaded areas represent the 95% CI of the median and the 5th and 95th percentiles of the simulated data, respectively.

Individual-predicted Post Hoc Exposure Metrics

- The exposures in the adolescent participants were generally within or slightly higher than the exposure range observed in the adult participants across all dosing phases (Figure 2).

Figure 2. Comparison of CAB Post Hoc Exposure in Adult and Adolescent Participants

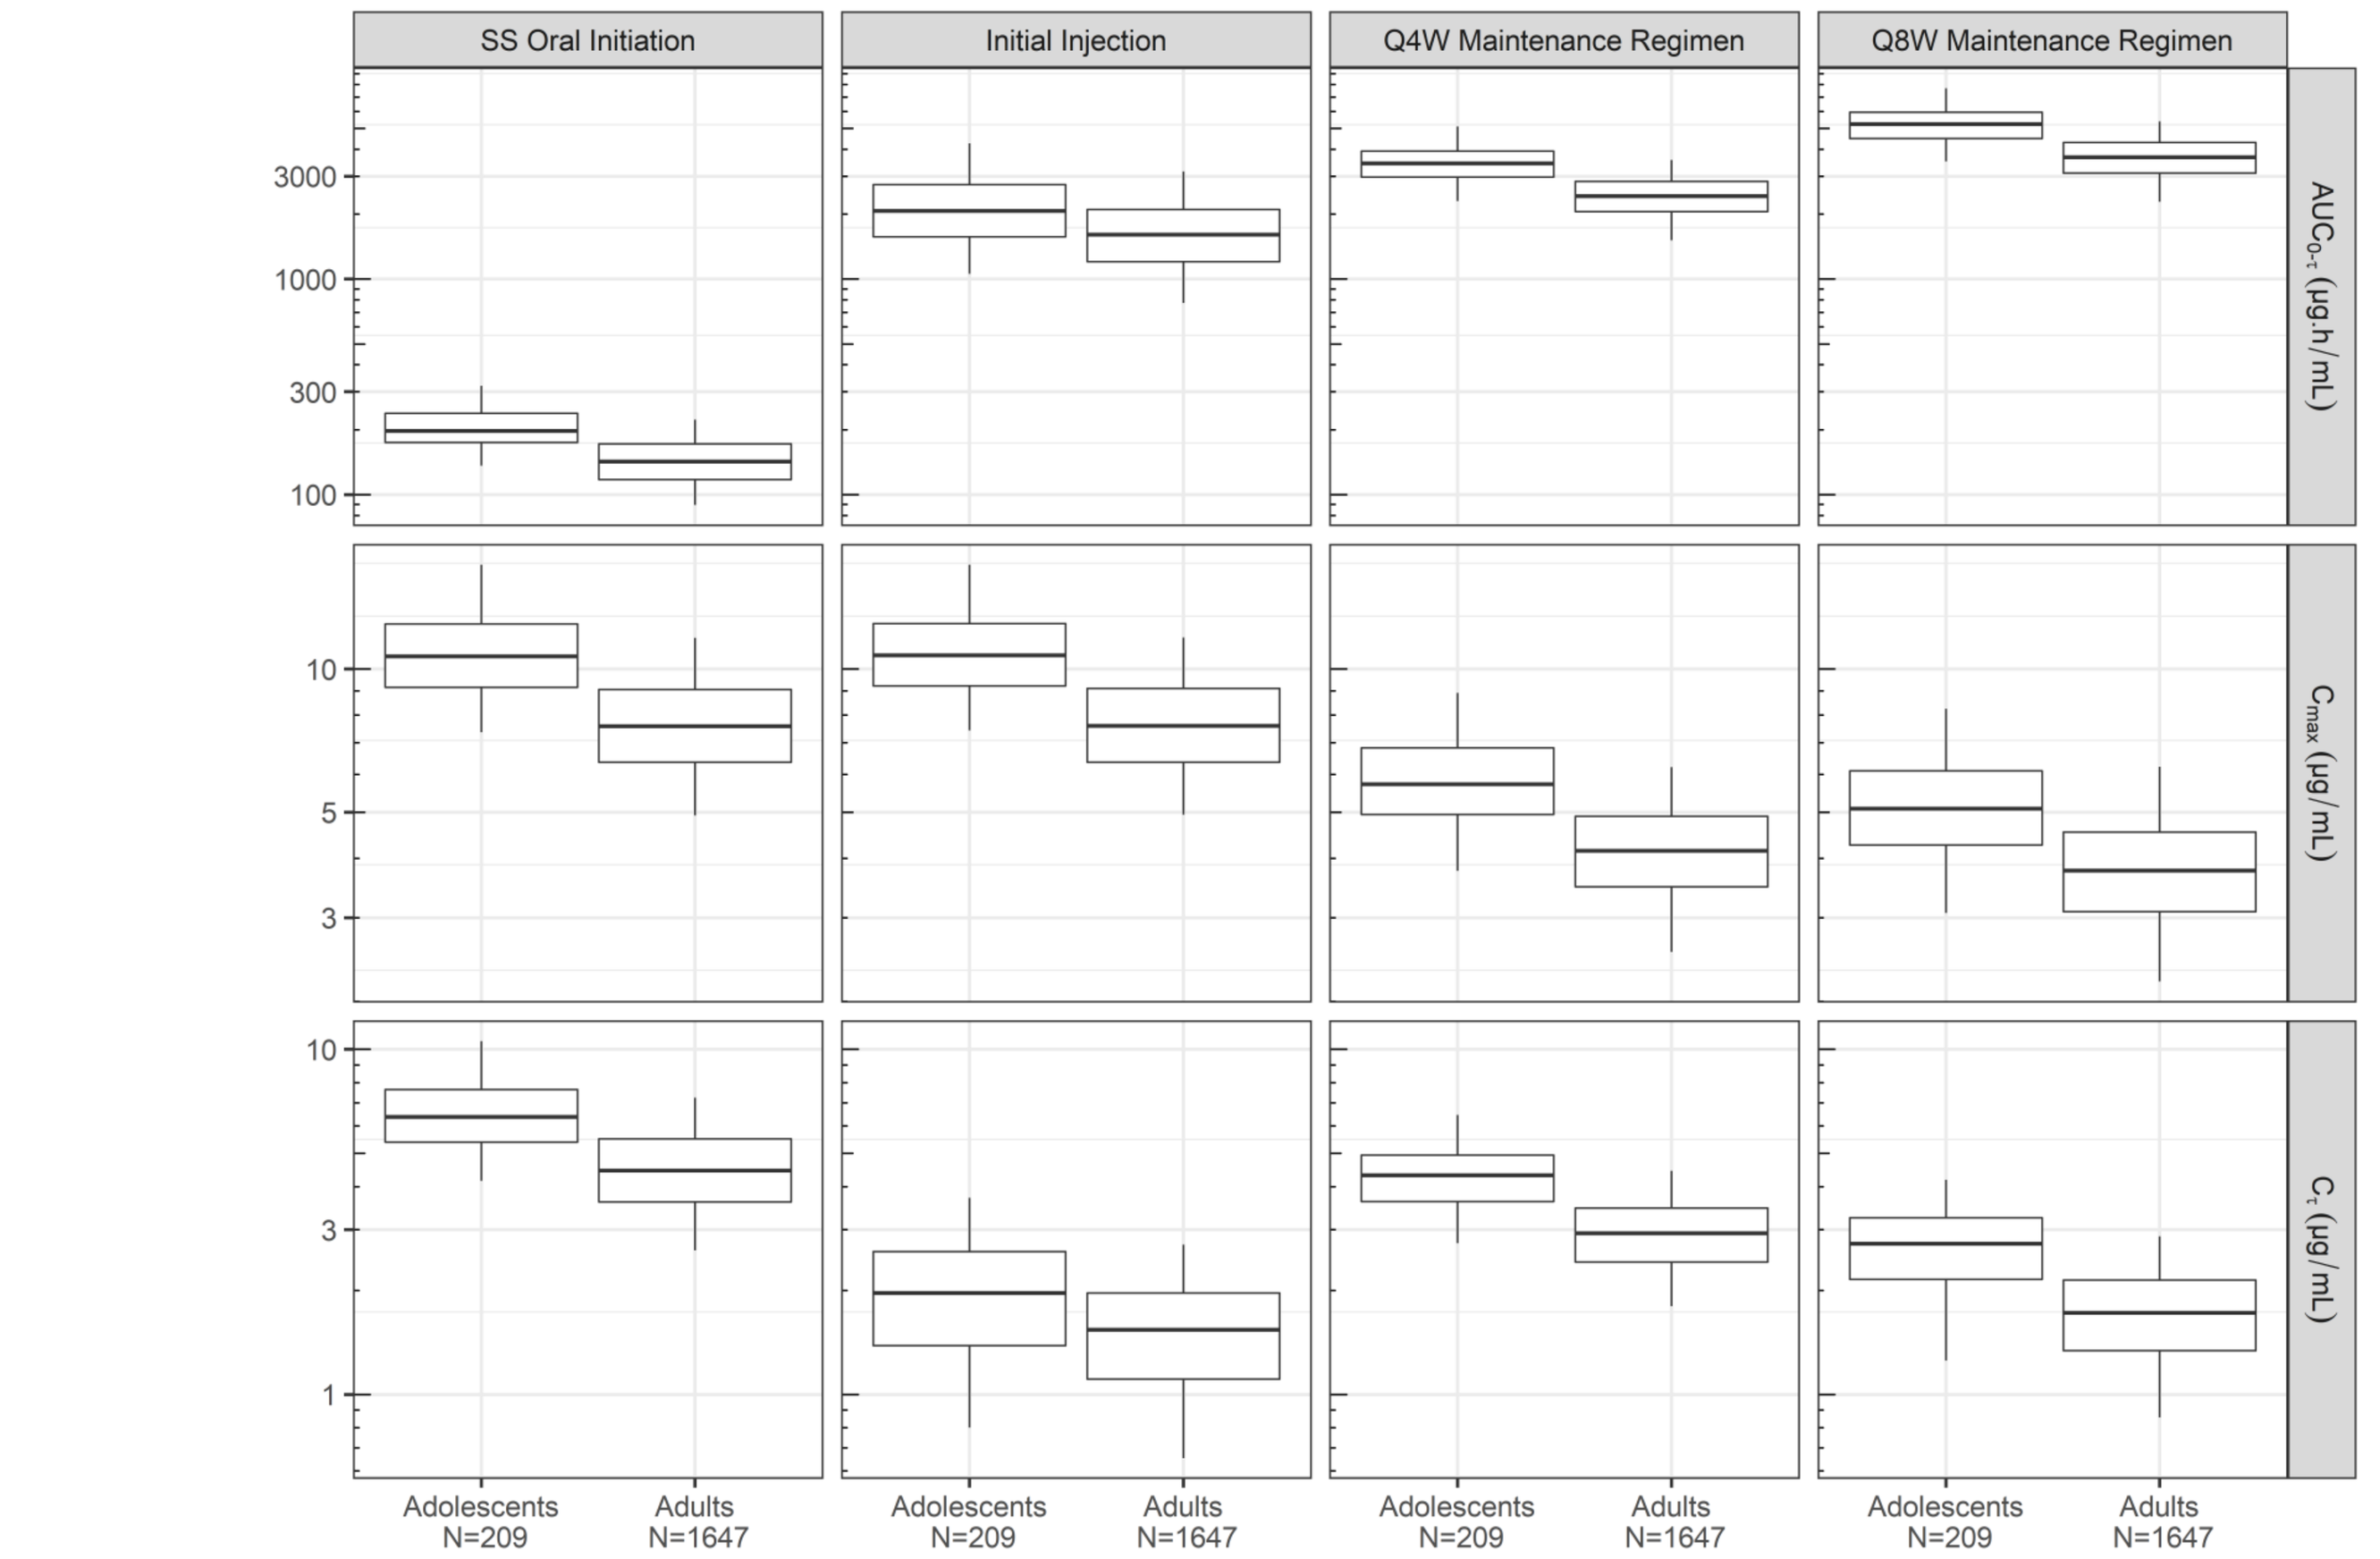
SS Oral Initiation
Initial Injection
Q4W Maintenance Regimen
Q8W Maintenance Regimen
AUC0-τ (µg.h/mL)
Cmax (µg/mL)
Cτ (µg/mL)
Adolescents N=209
Adults N=1647

Notes: The standard Q4W and Q8W dosing regimens were simulated with oral lead-in: oral lead-in of CAB PO QD for 4 weeks, followed by 600 mg CAB long-acting IM 2 hours after the last PO dose, followed by CAB long-acting IM 400 mg Q4W or 600 mg Q8W thereafter.
The Cmax following the first CAB LAI is likely determined by the last oral dose instead of the initiation long-acting dose.
For box plots, the horizontal center solid line in each box represents the median value, the box represents the 25th to 75th percentiles, and the whiskers represent the 5th and 95th percentiles. For AUC0-tau and Ctau, tau is different for different regimens.

Monte Carlo Simulations

- In various dosing scenarios (Q4W and Q8W dosing regimens), simulated concentrations were compared against the efficacy threshold of 0.45 µg/mL and the safety threshold of 22.5 µg/mL.[4]
- The CAB PK profiles across the entire treatment period for the CAB long-acting IM Q4W and Q8W dosing regimens were predicted to remain above the Phase 3 efficacy threshold and below the safety threshold in ≥95% of the virtual adolescent population (Figure 3).
- Adolescents had CAB LAI exposure at steady state ($C_{tau,ss}$ median, 5th-95th: **2.36, 0.849-4.13** µg/mL for 600 mg Q8W) comparable to that of adults ($C_{tau,ss}$: **1.91, 0.786-3.33** µg/mL for Q8W), with their exposure levels falling within the same range across all dosing phases, and contained within the established efficacy and safety thresholds (Figure 4).

Figure 3. CAB Concentration Versus Time Following Q4W/Q8W Dosing by Population

Q4W Standard Regimen | Q8W Standard Regimen

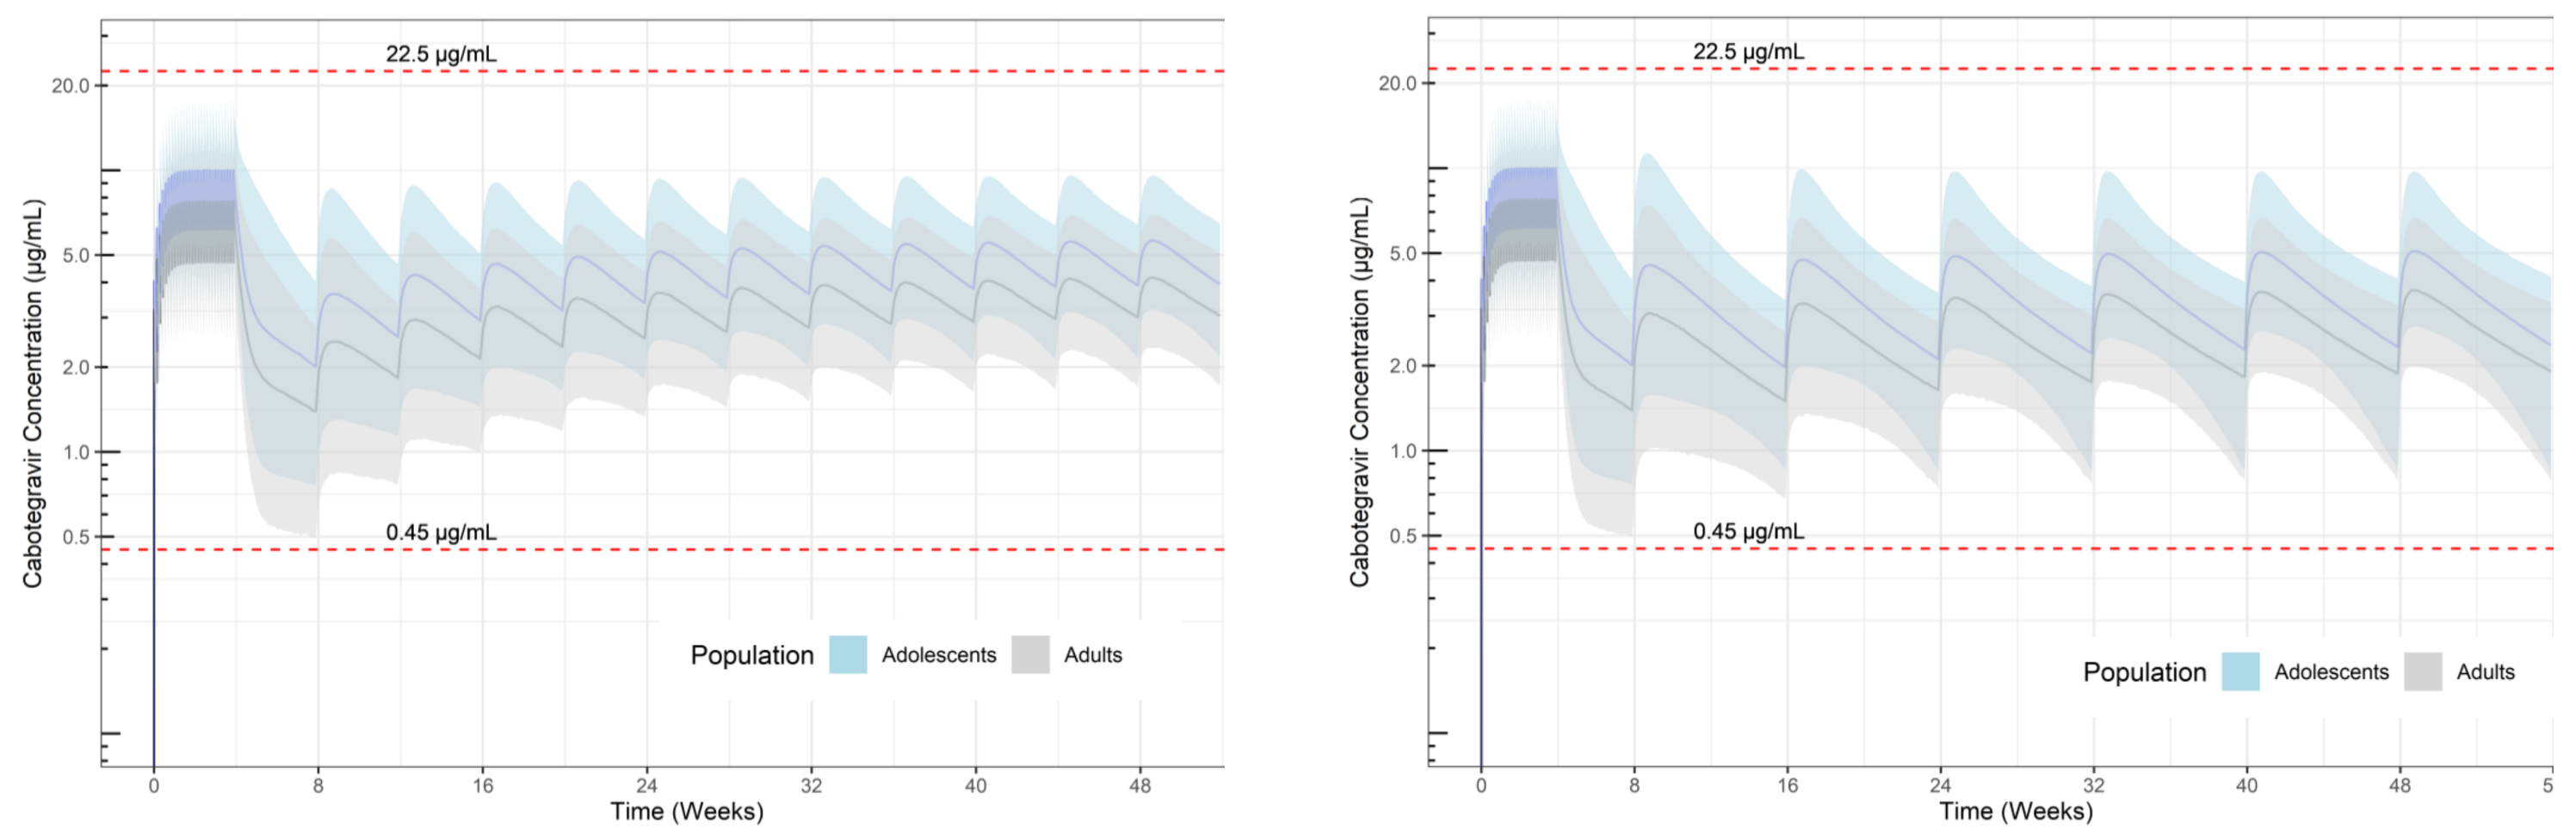
22.5 µg/mL
0.45 µg/mL
Cabotegravir Concentration (µg/mL)
Time (Weeks)
Population
Adolescents
Adults

Notes: The target outcome observed in participants is that 95% of participants who receive CAB should have trough concentrations during their treatment that exceed the phase 3 benchmark of 0.45 µg/mL. The short-term safety threshold of 22.5 µg/mL is an upper no-effect boundary for CAB, which represents the geometric mean of Cmax observed at the supratherapeutic dose of PO CAB 150 mg (3 doses in total, BID) in the thorough QT/QTc study (LAI117009).

Figure 4. Box Plot of C_{tau} Following Q4W and Q8W Dosing by Population

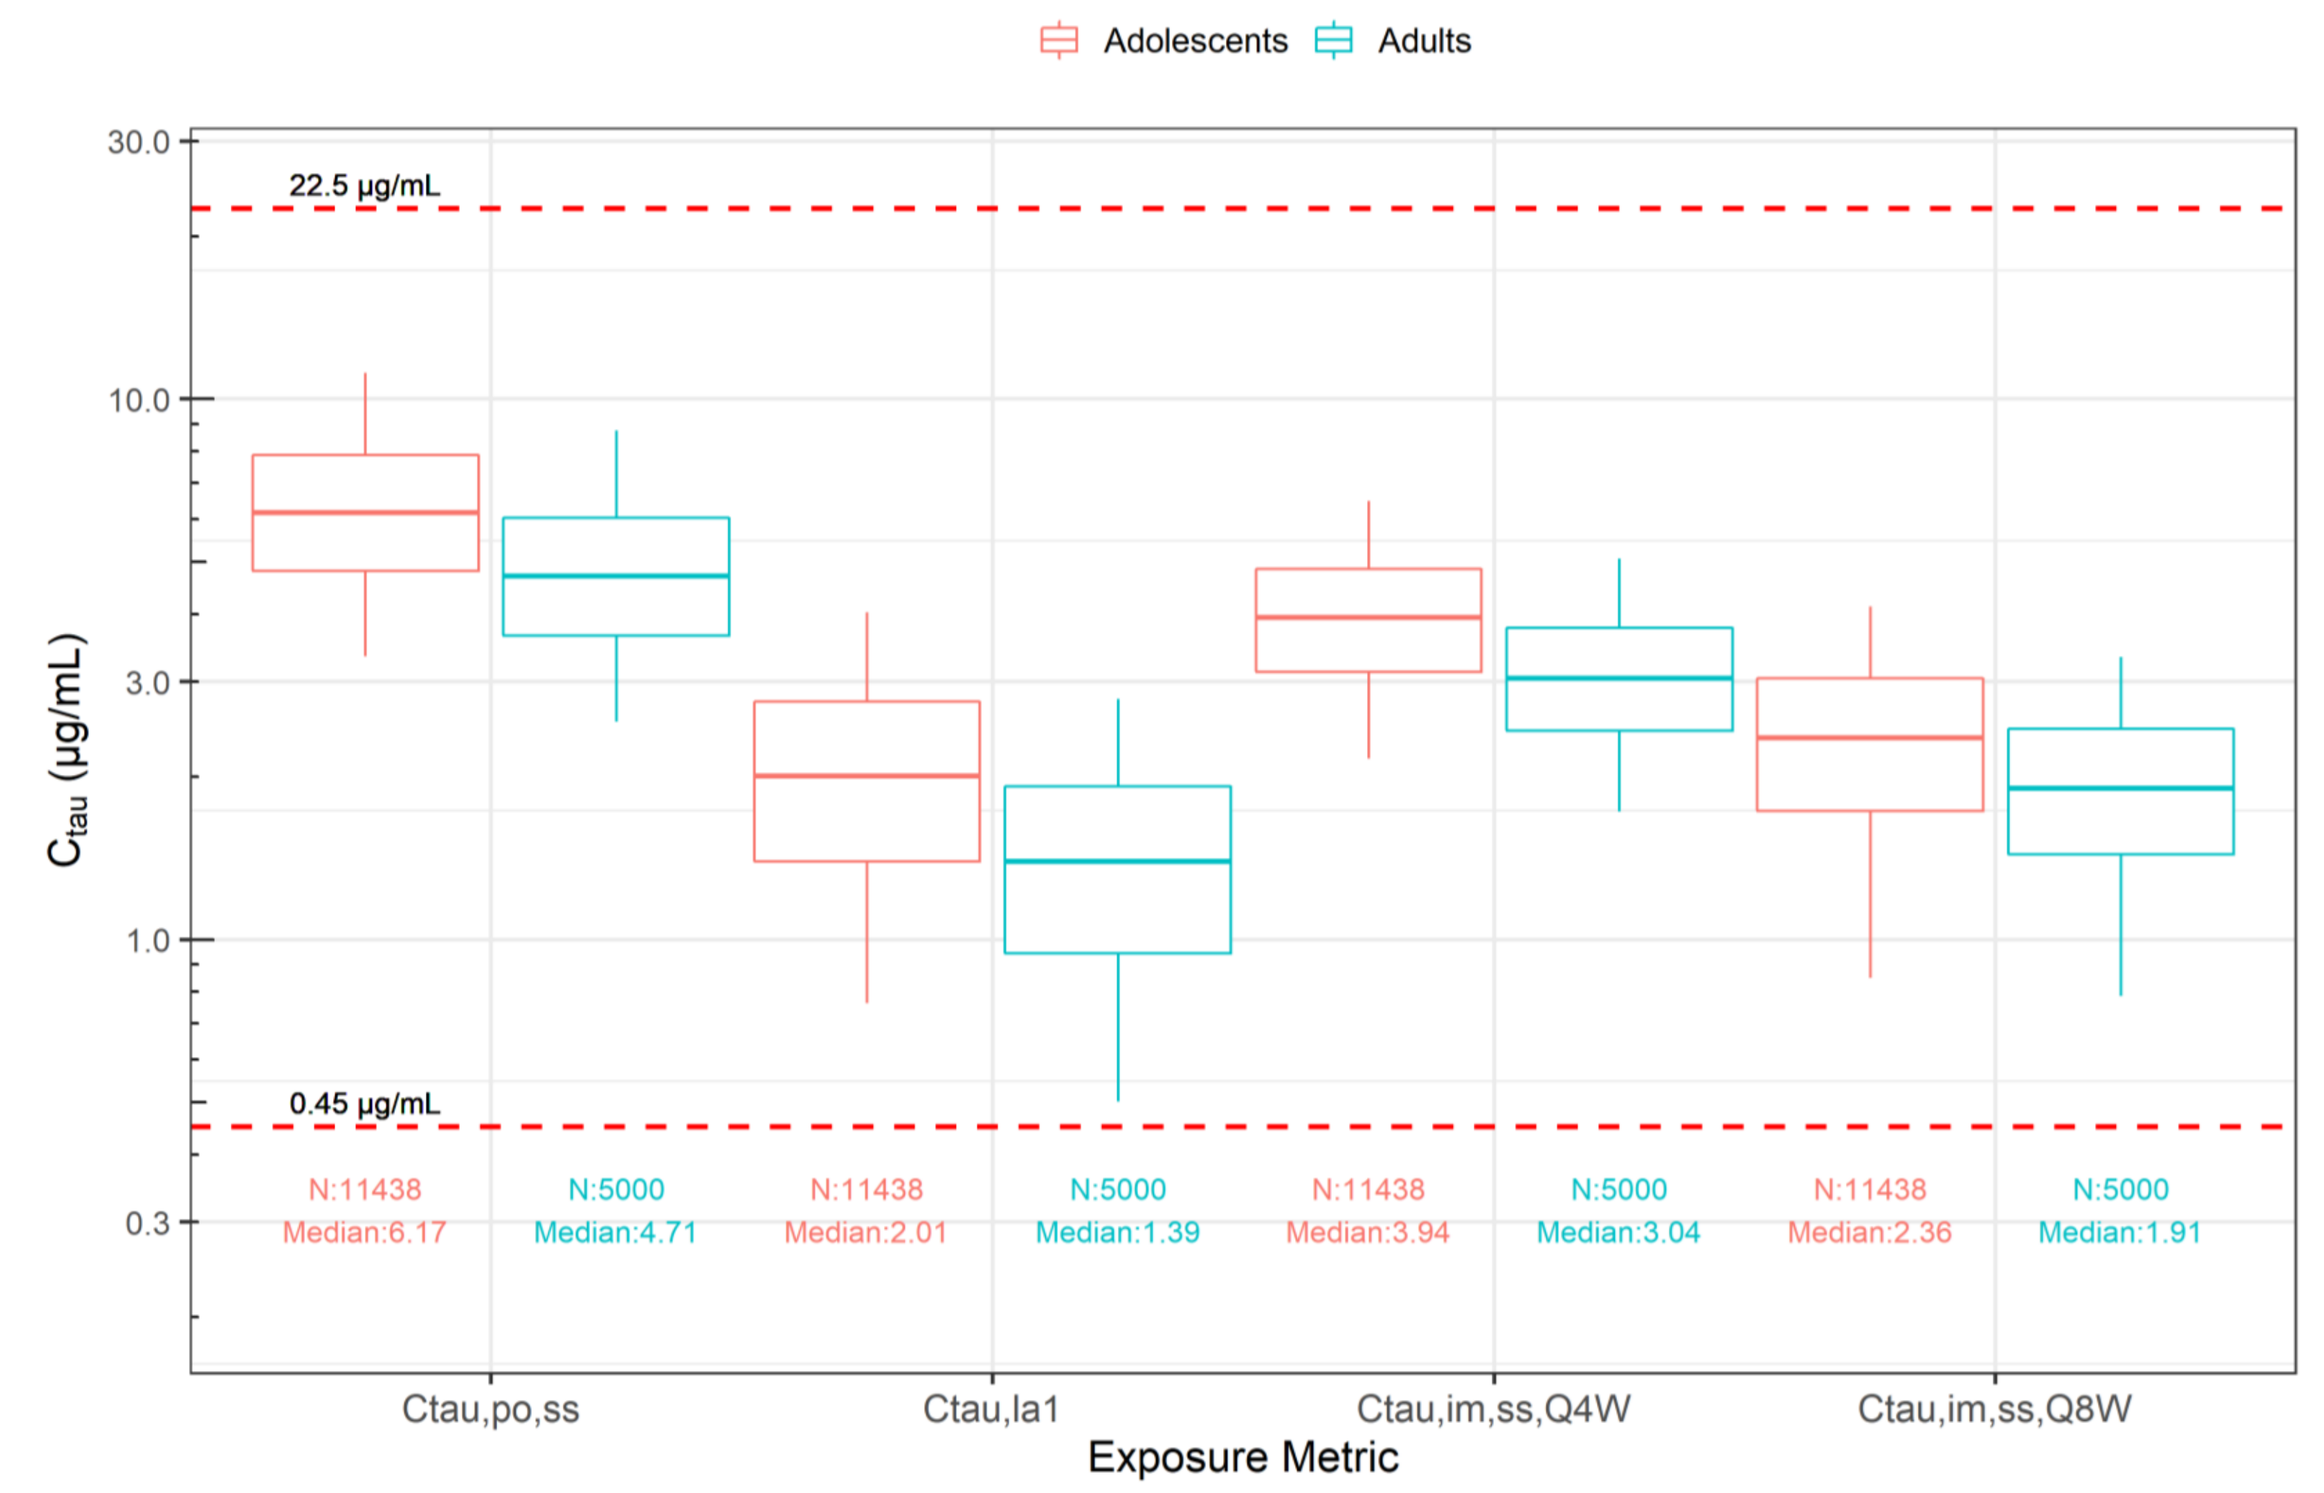
Adolescents
Adults
22.5 µg/mL
0.45 µg/mL
Ctau (µg/mL)
N:11438 Median:6.17
N:5000 Median:4.71
N:11438 Median:2.01
N:5000 Median:1.39
N:11438 Median:3.94
N:5000 Median:3.04
N:11438 Median:2.36
N:5000 Median:1.91
Ctau,po,ss
Ctau,la1
Ctau,im,ss,Q4W
Ctau,im,ss,Q8W
Exposure Metric

Notes: Ctau=plasma concentration at the end of the dosing interval; Ctau,im,ss=Ctau at steady state after the long-acting IM maintenance dose injections; Ctau,la1=Ctau at steady state after the first long-acting IM injection. The horizontal center solid line in each box represents the median value, the box represents the 25th to 75th percentiles, and the whiskers represent the 5th and 95th percentiles.

Conclusions

- The addition of adolescent data to the adult PopPK dataset allowed expansion of the prior PopPK model down to adolescents weighing 35 kg and optimization of predictions in adolescents.
- Given the similarity of CAB PK across adolescents and adults, the same dosing regimens apply for adults and adolescents. The slightly higher exposure in the adolescent participants is clinically insignificant.
- No dose adjustment is recommended for adolescent participants (12 to <18 years of age) weighing at least 35 kg in accordance with the current label for CAB.

Disclosure: Y-W.L, I.D. and S.Y.A.C are employees of Certara and own stocks in Certara. S.F is an employee of GSK and owns stock in GSK. J.C, C.M, and C.M.H are employees of ViiV Healthcare.
Overall support for the HIV Prevention Trials Network (HPTN) is provided by the National Institute of Allergy and Infectious Diseases (NIAID), Office of the Director (OD), National Institutes of Health (NIH), National Institute on Drug Abuse (NIDA), the National Institute of Mental Health (NIMH), and the Eunice Kennedy Shriver National Institute of Child Health and Human Development (NICHD) under Award Numbers UM1AI068619 (HPTN Leadership and Operations Center), UM1AI068617 (HPTN Statistical and Data Management Center), and UM1AI068613 (HPTN Laboratory Center).

References: 1. Han et al. *Br J Clin Pharmacol*. 2022;88(10):4607-4622. **2.** GSK Document No.: 2018N384611_01 (Population Pharmacokinetics Analysis of Cabotegravir. 2019). **3.** GSK Document No.: 2019N421460_00 (Population Pharmacokinetics Evaluation of Cabotegravir every 8 weeks Regimen. 2019). **4.** Integrated review. Page 86. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2021/212887Orig1s000,212888Orig1s000IntegratedR.pdf.

Conference on Retroviruses and Opportunistic Infections; March 3-6, 2024; Denver, CO

Presenting author: Susan Ford; Susan.l.ford@gsk.com